Safety, Tolerability, and Pharmacokinetics of Single and Multiple Doses of KNS-760704 in Healthy Volunteers

Cheryl F. Graham, MD,[1] James L. Mather,[1] Evan W. Ingersoll, Ph.D.,[1] William G. Kramer, Ph.D.,[2] Gregory T. Hebrank, MD, MBA,[1] and Michael E. Bozik, MD[1]

(1) Knopp Neurosciences Inc., 2100 Wharton Street, Suite 615, Pittsburgh, PA 15203
(2) Kramer Consulting LLC, 14313 Outpost Way, North Potomac, MD 20878

Abstract #037

ACCP Philadelphia; Sep 2008

INTRODUCTION

KNS 760704 [(6R)-4,5,6,7-tetrahydro-N6-propyl-2,6-benzothiazole-diamine dihydrochloride monohydrate] is a putative neuroprotective agent in clinical development for the treatment of patients with amyotrophic lateral sclerosis (ALS). It is the (R)-(+) optical enantiomer of the marketed compound pramipexole dihydrochloride (Mirapex®, Boehringer Ingelheim), a potent dopamine agonist with selective high affinity for the D2 and D3 receptor sub-types. Both enantiomers of pramipexole have been shown to possess neuroprotective properties. In contrast to pramipexole, KNS 760704 has very low affinities for D2 and D3 dopamine receptors and should be better tolerated at higher doses. The hypothesis for this clinical development program is that, when dosed at much higher levels than pramipexole, KNS 760704 may be an effective treatment for ALS. These two Phase 1 studies addressed the initial question of whether high doses are safe and well-tolerated in healthy volunteers.

OBJECTIVES

(1) To determine the safety and tolerability of ascending single and multiple doses of KNS 760704 in healthy volunteers, (2) to determine single- and multiple-dose pharmacokinetics, and (3) to preliminarily assess the effect of food on the absorption and elimination of KNS 760704.

METHODOLOGY

Single-Dose Study: KNS 760704-CL001 was a single-center, randomized, double-blind, placebo-controlled, ascending single-dose study. Safety observations included vital signs, physical examinations, clinical laboratory tests, electrocardiograms (ECGs), and adverse event (AE) reporting. Blood and urine samples were collected pre-dose and for 72 hours post-dose to assess the pharmacokinetics. A total of 30 subjects were enrolled in the study. Cohorts of eight subjects (six active and two placebo) were randomized to take placebo, KNS 760704 50 mg, KNS 760704 150 mg, or KNS 760704 300 mg in the fasted state. The 3 fasted dose panels were enrolled sequentially in an ascending dose manner and all available safety data were reviewed under blinded conditions before proceeding to the next dose level. A final panel of 6 subjects received KNS 760704 150 mg in the fed state. All subjects completed the study as planned.

Multiple-Dose Study: KNS 760704-CL002 was a single-center, randomized, double-blind, placebo-controlled, ascending multiple-dose study. The schedule of safety observations was the same as for the single-dose study. Cohorts of 8 subjects (6 active and 2 placebo) were administered a single dose of study drug or placebo on Day 1 followed 48 hours later by 4½ days of dosing every 12 hours (Q12H). Blood samples were collected pre-dose and serially for 48 hours post-dose on Day 1 to assess the single-dose pharmacokinetics. Blood samples were also collected pre-dose on Days 5, 6, and 7 and serially through 72 hours post-dose on Day 7 to assess the steady-state pharmacokinetics. Urine samples were collected for 12 hours after dosing on Day 7 to assess steady-state urinary excretion. Subjects were dosed under fasting conditions on the Day 1 and Day 7 pharmacokinetic days. A total of 24 healthy adult subjects were enrolled in 3 sequential panels to receive multiple doses of KNS 760704 50 mg Q12H (n=6), KNS 760704 100 mg Q12H (n=6), KNS 760704 Q12H 150 mg (n=6), or matching placebo Q12H (n=6). All subjects completed the study as planned.

RESULTS

Safety and Tolerability

The AE profile in each active dose group was similar to that in the placebo group. There was no evidence suggesting that specific treatment-emergent AEs by preferred term or by organ class were more frequent in subjects receiving KNS 760704 than in subjects receiving placebo. There were no clinically significant clinical tolerability or safety observations at any dose level, no serious adverse events or deaths, and no discontinuations due to AEs. Frequencies of AEs were similar between active- and placebo-treated groups (see Tables 1 and 2).

Table 1 (Single) **Number of Subjects with Adverse Events by System Organ Class and Preferred Term (Safety Population)**

System Organ Class / Preferred Term	Placebo (N=6)	KNS-760704 50 mg (N=6)	150 mg (N=6)	300 mg (N=6)	Fed 150 mg (N=6)
Subjects with ≥ 1 AE	4 (67%)	2 (33%)	3 (50%)	3 (50%)	3 (50%)
Cardiac disorders	1 (17%)	0	0	2 (33%)	1 (17%)
Tachycardia[a]	1 (17%)	0	0	2 (33%)	1 (17%)
Eye Disorders	0	0	0	0	1 (17%)
Visual disturbance	0	0	0	0	1 (17%)
Gastrointestinal disorders	0	0	1 (17%)	0	0
Nausea	0	0	1 (17%)	0	0
Vomiting	0	0	1 (17%)	0	0
General disorders and administration site conditions	0	0	0	0	1 (17%)
Sensation of pressure	0	0	0	0	1 (17%)
Investigations	0	0	0	1 (17%)	0
Blood creatinine increased[a,b]	0	0	0	1 (17%)	0
Blood triglycerides increased[a,b]	0	0	0	1 (17%)	0
Musculoskeletal and connective tissue disorders	3 (50%)	2 (33%)	0	0	1 (17%)
Muscle spasms	2 (33%)	2 (33%)	0	0	0
Back pain[b]	1 (17%)	0	0	0	0
Musculoskeletal stiffness	0	0	0	0	1 (17%)
Myalgia	0	1 (17%)	0	0	0
Nervous system disorders	3 (50%)	0	3 (50%)	1 (17%)	2 (33%)
Dizziness	3 (50%)	0	1 (17%)	1 (17%)	1 (17%)
Headache	1 (17%)	0	2 (33%)	0	1 (17%)
Psychiatric disorders	0	1 (17%)	0	0	0
Insomnia[b]	0	1 (17%)	0	0	0
Skin and subcutaneous tissue disorders	0	1 (17%)	0	0	1 (17%)
Hyperhidrosis	0	0	0	0	1 (17%)
Pruritus	0	1 (17%)	0	0	0

a Asymptomatic event identified by the Investigator
b Considered by the Investigator to be unlikely related or unrelated to study drug.

Table 2 (Multiple) **Number of Subjects with Adverse Events by System Organ Class and Preferred Term (Safety Population)**

System Organ Class / Preferred Term	Placebo (n=6)	KNS-760704 50 mg Q12H (n=6)	100 mg Q12H (n=6)	150 mg Q12H (n=6)
Subjects with ≥ 1 AE	3 (50%)	4 (67%)	6 (100%)	1 (17%)
Cardiac disorders	1 (17%)	1 (17%)	3 (50%)	1 (17%)
Palpitations	0	0	1 (17%)[a]	0
Tachycardia	1 (17%)[a]	1 (17%)[a]	2 (33%)[a]	1 (17%)[a]
Gastrointestinal disorders	0	0	1 (17%)	0
Dyspepsia	0	0	1 (17%)	0
General disorders and administration site conditions	0	1 (17%)	1 (17%)	0
Flushing	0	0	1 (17%)	0
Hot flush	0	1 (17%)	0	0
Hyperhidrosis	0	0	1 (17%)	0
Pallor	0	0	1 (17%)	0
Musculoskeletal and connective tissue disorders	1 (17%)	1 (17%)	2 (33%)	0
Back pain	1 (17%)	0	0	0
Musculoskeletal stiffness	0	0	1 (17%)	0
Pain in extremity	0	1 (17%)	1 (17%)	0
Nervous system disorders	1 (17%)	4 (67%)	2 (33%)	0
Dizziness	0	2 (33%)	2 (33%)	0
Headache	1 (17%)	4 (67%)	1 (17%)	0
Respiratory, thoracic and mediastinal disorders	0	0	1 (17%)	0
Sinus congestion	0	0	1 (17%)	0
Skin and subcutaneous tissue disorders	0	1 (17%)	0	0
Rash	0	1 (17%)	0	0

a Asymptomatic event reported by the Principal Investigator.

Figure 1. *Mean plasma KNS-760704 concentrations after oral administration of single 50 mg, 150 mg, and 300 mg doses to adult volunteers under fasted conditions. Semi-logarithmic axes.*

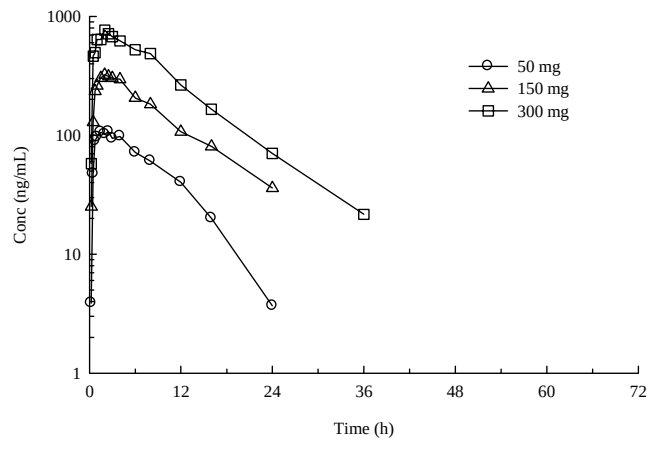
1000
100
10
1
Conc (ng/mL)
0
12
24
36
48
60
72
Time (h)
50 mg
150 mg
300 mg

Figure 2. *Mean Plasma KNS-760704 Concentrations after Oral Administration of Single 150 mg Doses to Adult Subjects under Fasted and Fed Conditions — Semi-logarithmic Axes*

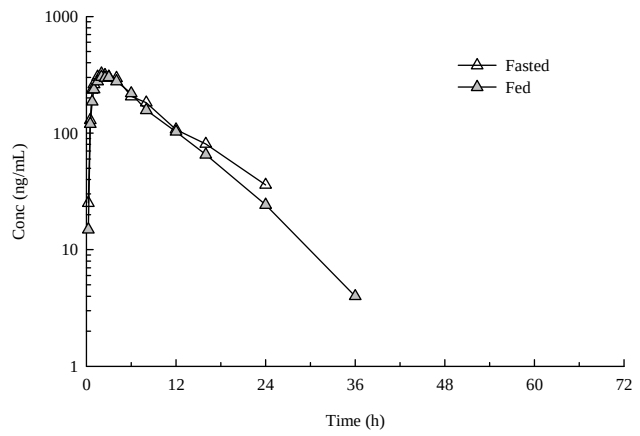
1000
100
10
1
Conc (ng/mL)
0
12
24
36
48
60
72
Time (h)
Fasted
Fed

Single-Dose Pharmacokinetics

Single-dose pharmacokinetics of KNS 760704 was linear over the doses studied with rapid absorption (Tmax ~2 h) with mean maximum concentrations (Cmax) of 125, 360, and 781 ng/mL and mean exposures (AUC0-∞) of 1254, 3782, and 8624 h×ng/mL for the 50, 150, and 300 mg single-dose groups, respectively (Table 3 and Figure 1). Urinary excretion of unchanged drug accounted for 90% of the dose. Both Cmax and AUC increased in proportion to dose across the dose levels tested. The mean T½ was independent of dose and ranged from 6.40 hours to 6.96 hours across the 3 fasted dose levels. CLr was ~4- to 5-fold greater than the glomerular filtration rate of ~120 mL/min (Table 3), indicating that active tubular secretion is likely a substantial component of the renal excretion of KNS 760704. It did not appear to be saturated at the doses administered. There was no effect of food on the absorption or elimination of KNS 760704 (see Figure 2).

Table 3. *Summary of Pharmacokinetic Parameters for KNS-760704 after Oral Administration of Single 50 mg, 150 mg, and 300 mg Doses under Fasted Conditions and 150 mg under Fed Conditions*

Parameter[1]	Fasted 50 mg	Fasted 150 mg	Fasted 300 mg	Fed 150 mg
Cmax (ng/mL)	125 ± 22.0 (6)	360 ± 60.4 (6)	781 ± 158 (6)	315 ± 062 (6)
Tmax (h)	2.04 (6)	2.04 (6)	1.98 (6)	2.58 (6)
AUC(0-t) (h•ng/mL)	989 ± 295 (6)	3,360 ± 780 (6)	8,340 ± 3,203 (6)	3,080 ± 934 (6)
AUC(inf) (h•ng/mL)	1,254 ± 347 (6)	3,782 ± 1,012 (5)	8,624 ± 3,263 (6)	3,379 ± 957 (6)
λz (h^{-1})	0.1064 ± 0.0171 (6)	0.1001 ± 0.0087 (5)	0.1151 ± 0.0309 (6)	0.1144 ± 0.0259 (6)
t½ (h)	6.65 ± 1.07 (6)	6.96 ± 0.56 (5)	6.40 ± 1.73 (6)	6.33 ± 1.49 (6)
CL/F (mL/min)	527 ± 135 (6)	524 ± 146 (5)	492 ± 194 (6)	581 ± 127 (6)
Vz/F (L)	294 ± 46.2 (6)	311 ± 68.4 (5)	258 ± 73.5 (6)	308 ± 55.9 (6)
Ue (mg)	35.3 ± 5.19 (6)	57.0 ± 12.6 (6)	198 ± 28.0 (6)	96.9 ± 4.71 (6)
Fe (% Dose)	94.7 ± 13.9 (6)	51.0 ± 11.2 (6)	88.3 ± 12.5 (6)	86.6 ± 4.21 (6)
CLr (mL/min)	628 ± 149 (6)	304 ± 122 (6)	441 ± 159 (6)	559 ± 140 (6)

Multiple-Dose Pharmacokinetics

Multiple-dose PK was also linear, with Cmax of 191, 306, and 479 ng/mL and AUC(0-12) of 1449, 2467, and 3749 h×ng/mL for the 50, 100, and 150 mg Q12H groups, respectively following 4½ days of twice-daily dosing (Table 4 and Figure 3). Tmax and urinary excretion following multiple doses were similar to the single-dose results. The accumulation was 1.2-fold to 1.4-fold, consistent with the half-life and dosing interval and further supports the linearity of the pharmacokinetics. The elimination half-life was approximately 7.6 hours, and approximately 84% of the dose was recovered in the urine over a 12-hour steady-state dosing period. Renal clearance is much greater than glomerular filtration, consistent with active secretion, but does not appear to be saturated at the doses administered.

Table 4. *Summary of Pharmacokinetic Parameters for KNS-760704 on Day 7 during Oral Administration of 50 mg, 100 mg, and 150 mg doses on Day 1, Q12H on Days 3 through 6, and a Single Dose on Day 7 to Healthy Volunteers under Fasted Conditions*

Parameter[1]	Dose 50 mg Q12H	100 mg Q12H	150 mg Q12H
Cmax (ng/mL)	191 ± 20.9 (6)	306 ± 54.8 (6)	479 ± 74.6 (6)
Tmax (h)	1.75 (6)	2.02 (6)	2.18 (6)
AUC(0-12) (h•ng/mL)	1,449 ± 221 (6)	2,467 ± 304 (6)	3,749 ± 575 (6)
λz (h^{-1})	0.1039 ± 0.0193 (6)	0.0893 ± 0.0117 (6)	0.0895 ± 0.0184 (6)
t½ (h)	6.87 ± 1.29 (6)	7.89 ± 1.19 (6)	8.05 ± 1.80 (6)
CL/F (mL/min)	437 ± 60.8 (6)	510 ± 57.4 (6)	507 ± 74.1 (6)
Vz/F (L)	255 ± 24.0 (6)	348 ± 61.7 (6)	349 ± 75.8 (6)
Ue (mg)	32.5 ± 3.82 (6)	56.3 ± 2.62 (4)	100 ± 8.76 (5)
Fe (% Dose)	87.2 ± 10.3 (6)	75.5 ± 3.51 (4)	89.6 ± 7.83 (5)
CLr (mL/min)	385 ± 90.3 (6)	382 ± 62.7 (4)	451 ± 102 (5)

[1] Mean ± standard deviation (N) except for Tmax for which the median (N) is reported.

Figure 3. *Mean plasma KNS 760704 concentrations on Day 7 during oral administration of single 50 mg, 100 mg, and 150 mg doses on Day 1, Q12H doses on Days 3 through 6, and single doses on Day 7 to healthy volunteers under fasted conditions — semi-logarithmic axes.*

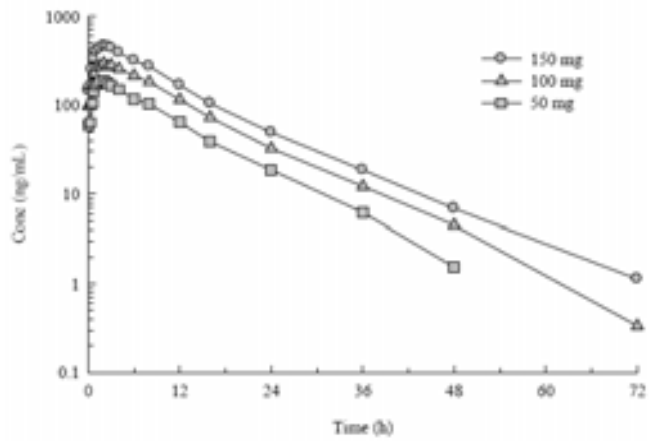
1000
100
10
1
0.1
Conc (ng/mL)
0
12
24
36
48
60
72
Time (h)
150 mg
100 mg
50 mg

CONCLUSIONS

These data demonstrate that KNS 760704 is safe and well tolerated with linear PK at single doses up to 300 mg and multiple doses up to 150 mg Q12H for 4½ days. Renal clearance at steady state is comparable to that following single doses indicating that renal clearance is not saturated at a 300 mg/day total daily dose.